NLX-112 has favorable safety, tolerability and efficacy against levodopa-induced dyskinesia (LID) in a randomized, double-blind, placebo-controlled, proof-of-concept Ph2A study

P. Svenningsson[1], P. Odin[2], F. Bergquist[3], K. Wirdefeldt[4], D. Nyholm[5], M. Andréasson[1], I. Markaki[1], A. Johansson[4], M. Jergil[6], C. Jankosky[7], M.A. Varney[7], F. Herbrecht[7], S.A. Johnson[7], A. Newman-Tancredi[7]

[1] ASC Torsplan, Stockholm, Sweden; [2] Skåne University Hospital, Lund, Sweden; [3] Sahlgrenska Hospital, Gothenburg, Sweden; [4] Karolinska University Hospital, Stockholm, Sweden; [5] Uppsala University, Sweden; [6] CTC-AB, 752 37 Uppsala, Sweden; [7] Neurolixis, 81100 Castres, France

Introduction

Levodopa-induced-dyskinesia (LID) in People with Parkinson (PwP) is a troublesome side-effect of extended (5-10 years) treatment with levodopa. LID is thought to result from the uptake of levodopa by 5-hydroxytryptamine (5-HT) neurons and its conversion to dopamine (DA), resulting in non-physiological release of DA (the "false neurotransmitter" effect). This leads to dyskinesia (erratic involuntary face, trunk or limb movements) which is often managed by reducing levodopa doses, and/or co-treatment with amantadine, the only approved drug for LID.

However, amantadine is associated with unwanted side-effects such as hallucinations and confusion as well as cardiovascular and gastrointestinal effects. Moreover, a substantial proportion of PwP either do not respond or experience a loss of anti-LID efficacy over time.

NLX-112 (a.k.a. befiradol), is a novel chemical entity which is centrally acting and is exceptionally selective for serotonin 5-hydroxytryptamine 1A (5-HT$_{1A}$) receptors. It is very potent (nanomolar binding affinity) and has high-efficacy 'full agonist' properties [1].

In 6-OH-DA-lesioned rats and MPTP-treated marmosets and cynomolgus macaques subjected to a chronic regimen of levodopa to trigger dyskinesia, NLX-112 potently and efficaciously reduced Abnormal Involuntary Movements (AIMs) and dyskinesia scores, preclinical measures predictive of anti-LID activity [2, 3, 4, 5].

NLX-112 has previously been shown to exhibit favorable safety and tolerability in clinical trials for an unrelated disorder (over 600 subjects exposed).

The present trial was supported by Parkinson's UK (DYS-20-02) and The Michael J. Fox Foundation (MJFF- 019508).

Methods

NLX-112 was tested in a randomized, double-blind, placebo (PBO)-controlled phase 2A proof-of-concept (POC) trial under Good Clinical Practice. The study consisted of 8-weeks of total treatment at 5 clinical sites in Sweden (ClinicalTrials.gov ID # NCT05148884).

PwP with troublesome LID received NLX-112 or PBO (2:1 ratio) as an adjunct to their regular stable antiparkinsonian medication. Study drug dosing was individually up-titrated over 28 days to a maximum of 2 mg/day (1 mg b.i.d.); dosing was then kept stable for 14 days and down-titrated over the next 14 days.

Subjects received a levodopa challenge (150% of regular dose) at day 1 (baseline) and at test days 28 and 42 to increase uniformity of LIDs and reduce day-to-day variability. The primary outcome was safety and tolerability; secondary outcome was change from baseline in troublesome peak LID as determined using the United Dyskinesia Rating Scale (UDysRS). Parkinsonism was assessed using the United Parkinson's Disease Rating Scale (UPDRS). Other motor and non-motor symptoms of PD were also evaluated.

Timeline and Office Visits

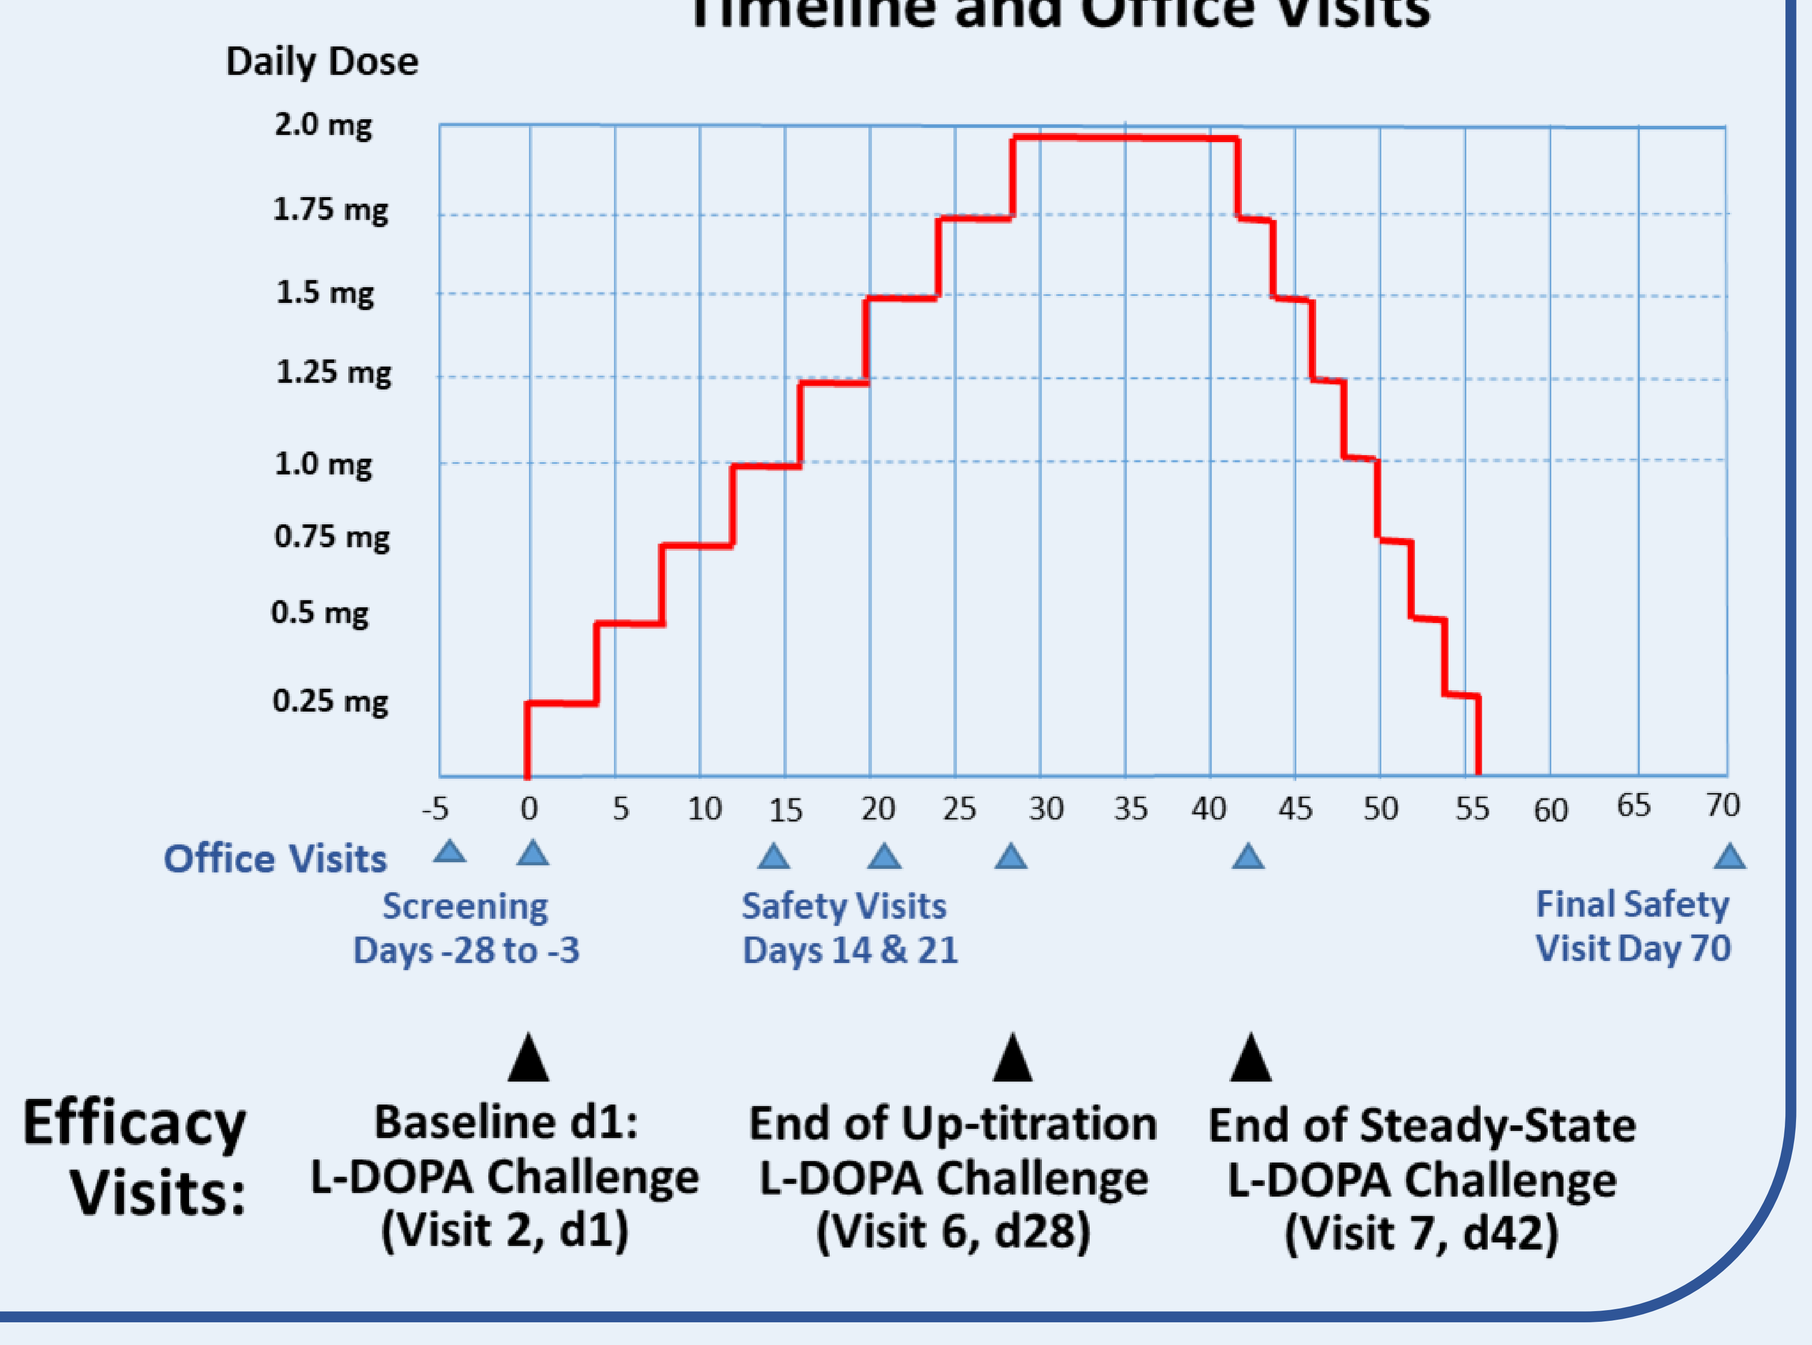
Daily Dose
2.0 mg
1.75 mg
1.5 mg
1.25 mg
1.0 mg
0.75 mg
0.5 mg
0.25 mg
-5 0 5 10 15 20 25 30 35 40 45 50 55 60 65 70
Office Visits
Screening Days -28 to -3
Safety Visits Days 14 & 21
Final Safety Visit Day 70
Efficacy Visits:
Baseline d1: L-DOPA Challenge (Visit 2, d1)
End of Up-titration L-DOPA Challenge (Visit 6, d28)
End of Steady-State L-DOPA Challenge (Visit 7, d42)

Subject demographics and baseline characteristics

Assessment	Unit	NLX-112 (N=18)	Placebo (N=9)
Age	Years, mean (SD)	65.7 (9.7)	64.6 (6.3)
Sex	'n' (male %)	10/18 (56%)	5/9 (56%)
Ethnicity	Hispanic or latino	1/18 (5.6%)	0
	Not hispanic or latino	17/18 (94%)	9/9 (100%)
Race	'n' (white %)	17/18 (94%)	9/9 (100%)
Time since diagnosis	Years, mean (SD)	11.4 (4.2)	9.9 (4.6)
Hoehn & Yahr score	Mean (SD)	2.5 (0.6)	2.6 (0.5)

Subject Disposition

Screened (N=35)

Randomized (N=27)

NLX-112 (N=18) — Discontinuations: Withdrawal of consent (N=3) — **Completed** (N=15)

Placebo (N=9) — Discontinuation: DBS Surgery (N=1) — **Completed** (N=8)

23 subjects completed the study. One subject in the placebo group was excluded (protocol infringement). The Per-Protocol Set (PPS) consisted of 22 subjects.

1) NLX-112 was safe and generally well tolerated

	NLX-112 (N=18)	Placebo (N=9)
Total AEs	16 (89%)	7 (78%)
AEs leading to withdrawal	1 (5.6%)	0
Severity:		
Mild	14 (78%)	7 (78%)
Moderate	10 (56%)	4 (44%)
Severe	0	1 (11%)

- Similar number of AEs were reported by subjects receiving placebo and NLX-112
- Most AEs were observed during up-titration
- All AEs in the NLX-112 group were either mild or moderate
- No SAEs were reported in the NLX-112 group (one SAE in the placebo group).

Preferred Term	NLX-112 (N=18)	Placebo (N=9)
Nausea	4 (22%)	0 (0%)
Headache	3 (17%)	2 (22%)
Parkinsonism	3 (17%)	3 (33%)
Insomnia	2 (11%)	0 (0%)
Vomiting	2 (11%)	1 (11%)
Fatigue	2 (11%)	2 (22%)
Orthostatic hypotension	2 (11%)	1 (11%)
Dizziness	2 (11%)	1 (11%)
Vertigo	2 (11%)	1 (11%)
Fall	2 (11%)	0 (0%)
Back pain	2 (11%)	0 (0%)
Restless leg syndrome	2 (11%)	0 (0%)
Dissociation	2 (11%)	0 (0%)

2) NLX-112 significantly reduced peak LID

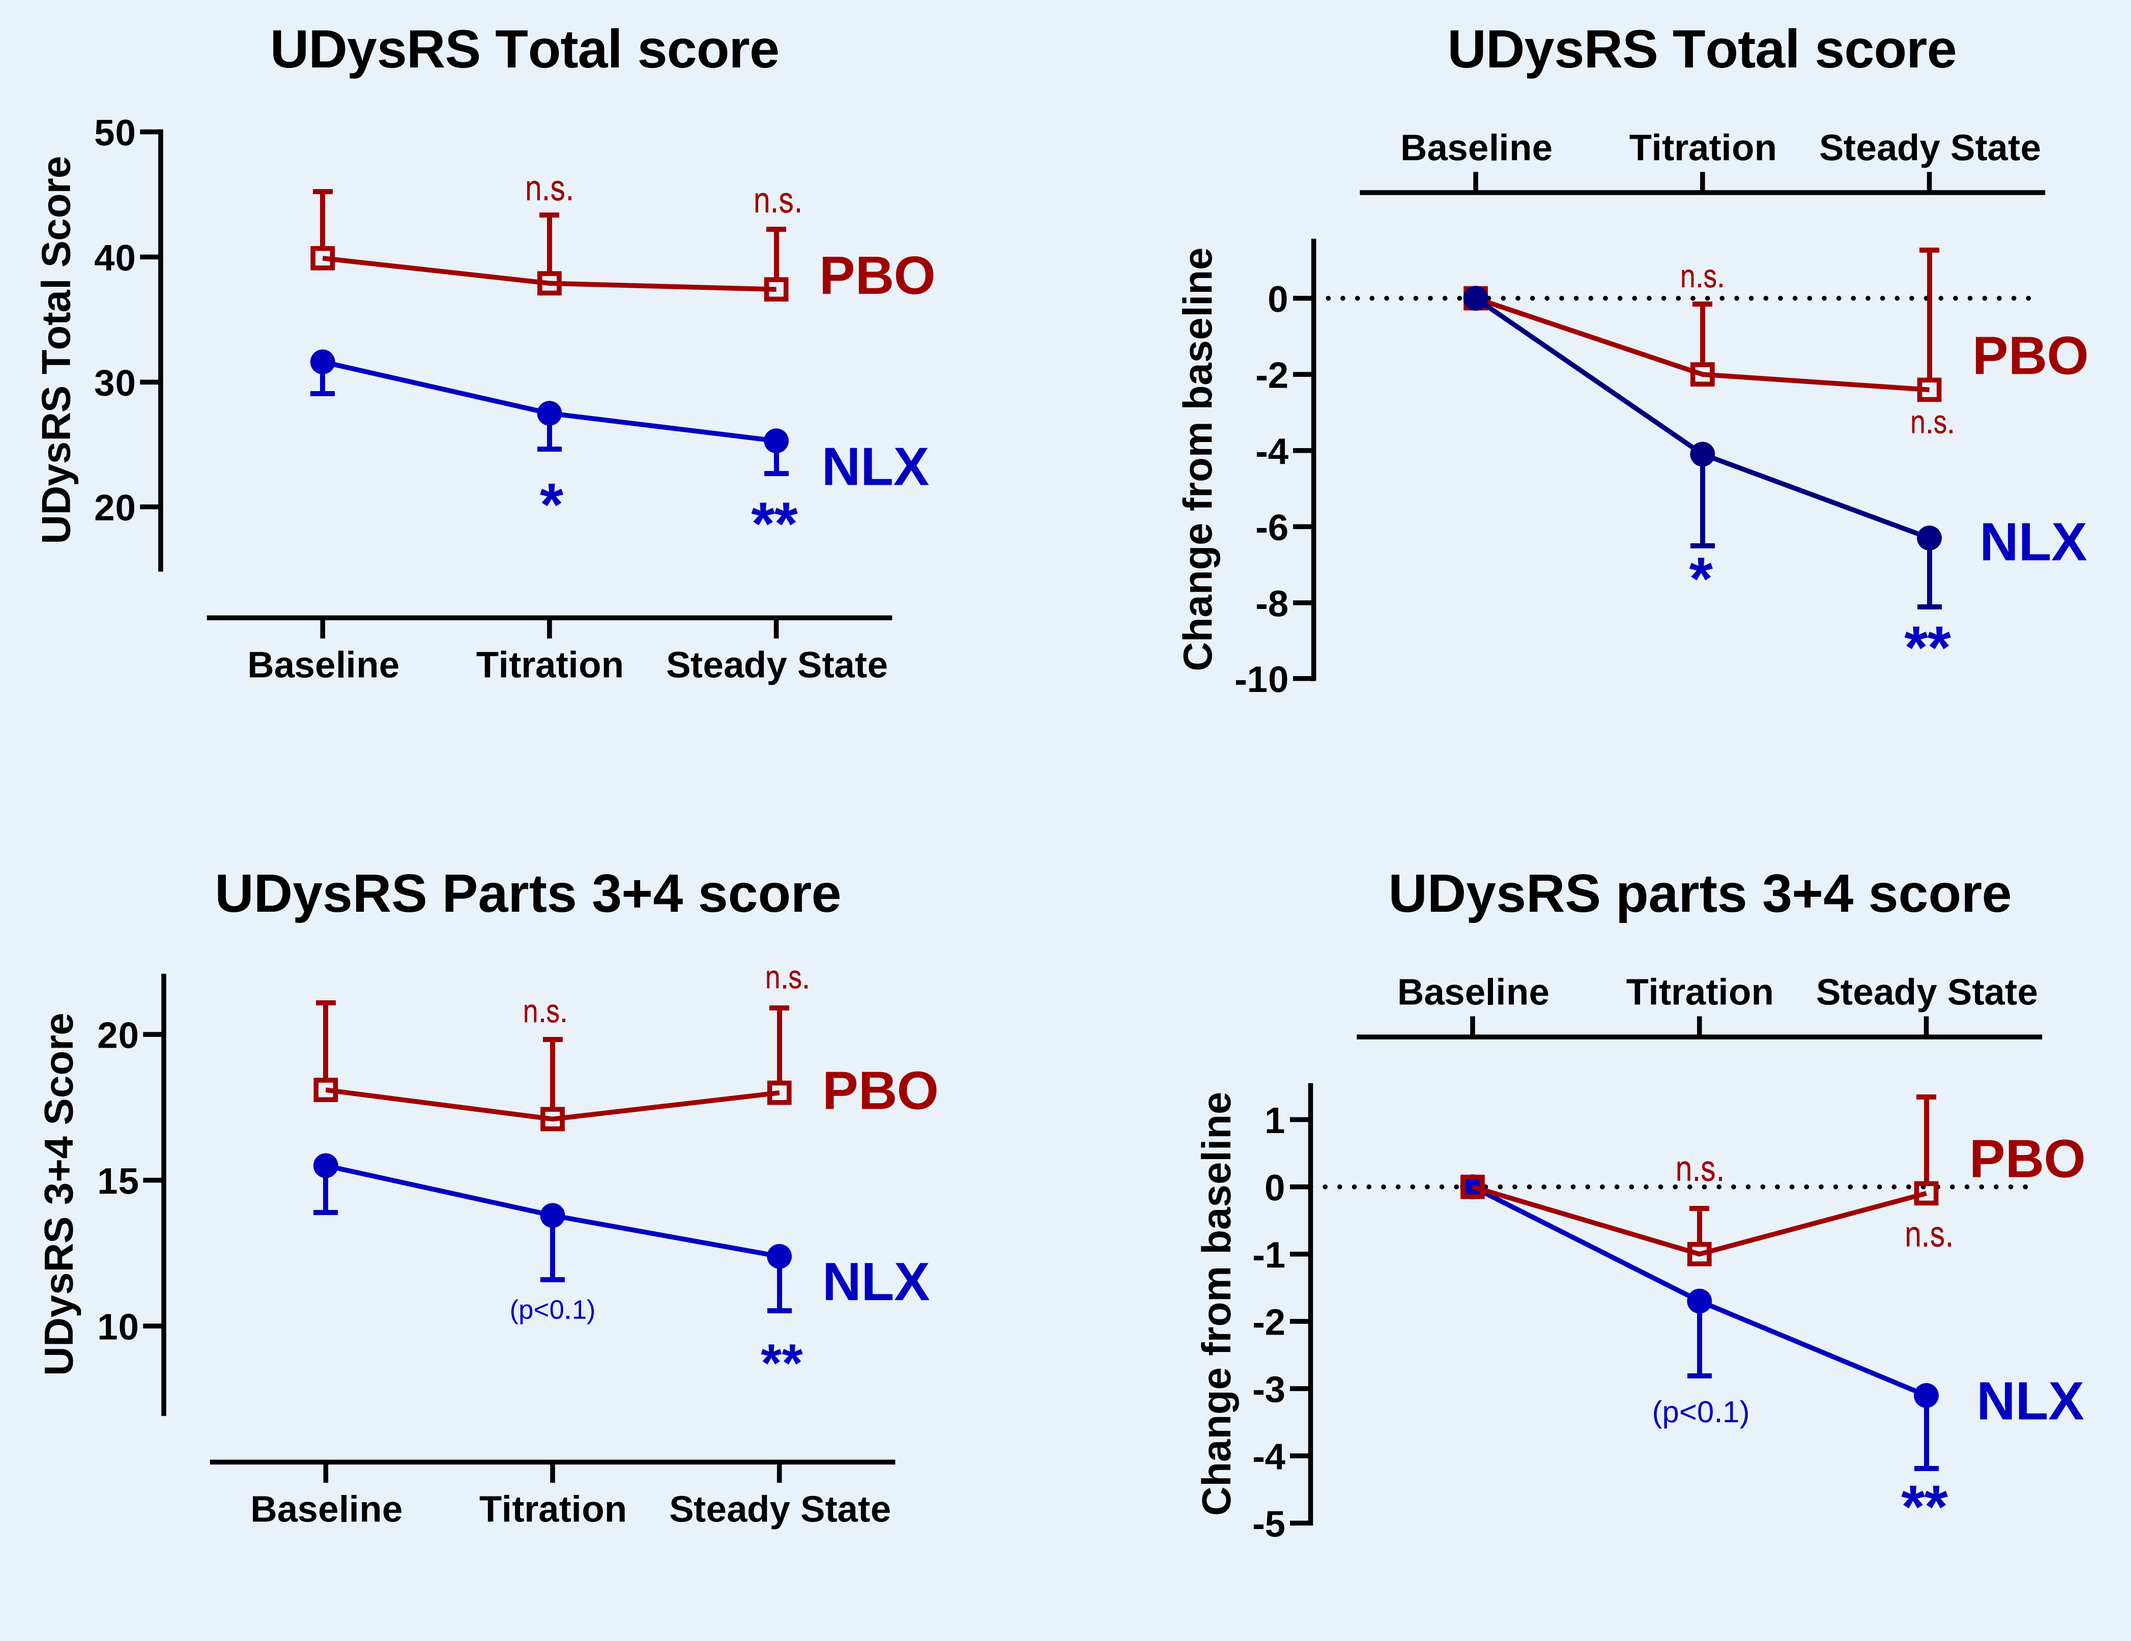
UDysRS Total score
UDysRS Total Score
PBO
NLX
Baseline Titration Steady State
UDysRS Total score
Change from baseline
UDysRS Parts 3+4 score
UDysRS 3+4 Score
(p<0.1)
UDysRS parts 3+4 score

At day 28 (end of up-titration), in the NLX-112 group, UDysRS total score (parts 1-4) decreased by-4.1 points ($p=0.0281$) and part 3+4 score (dyskinesia disability) by -1.7 points (n.s.) compared to baseline; placebo (PBO) group changes were n.s. (-2.0 and -1.0, respectively).

At day 42 (end of steady state period), a greater reduction of UDysRS scores was observed in the NLX-112 group: total score decreased by -6.3 points ($p=0.0016$) and part 3+4 score by -3.1 ($p=0.0038$) compared to baseline; PBO group changes were n.s. (-2.4 and -0.1, respectively).

***** p <0.05 vs baseline; ****** p< 0.01 vs baseline; n.s. not significant

3) NLX-112 significantly further reduced parkinsonism

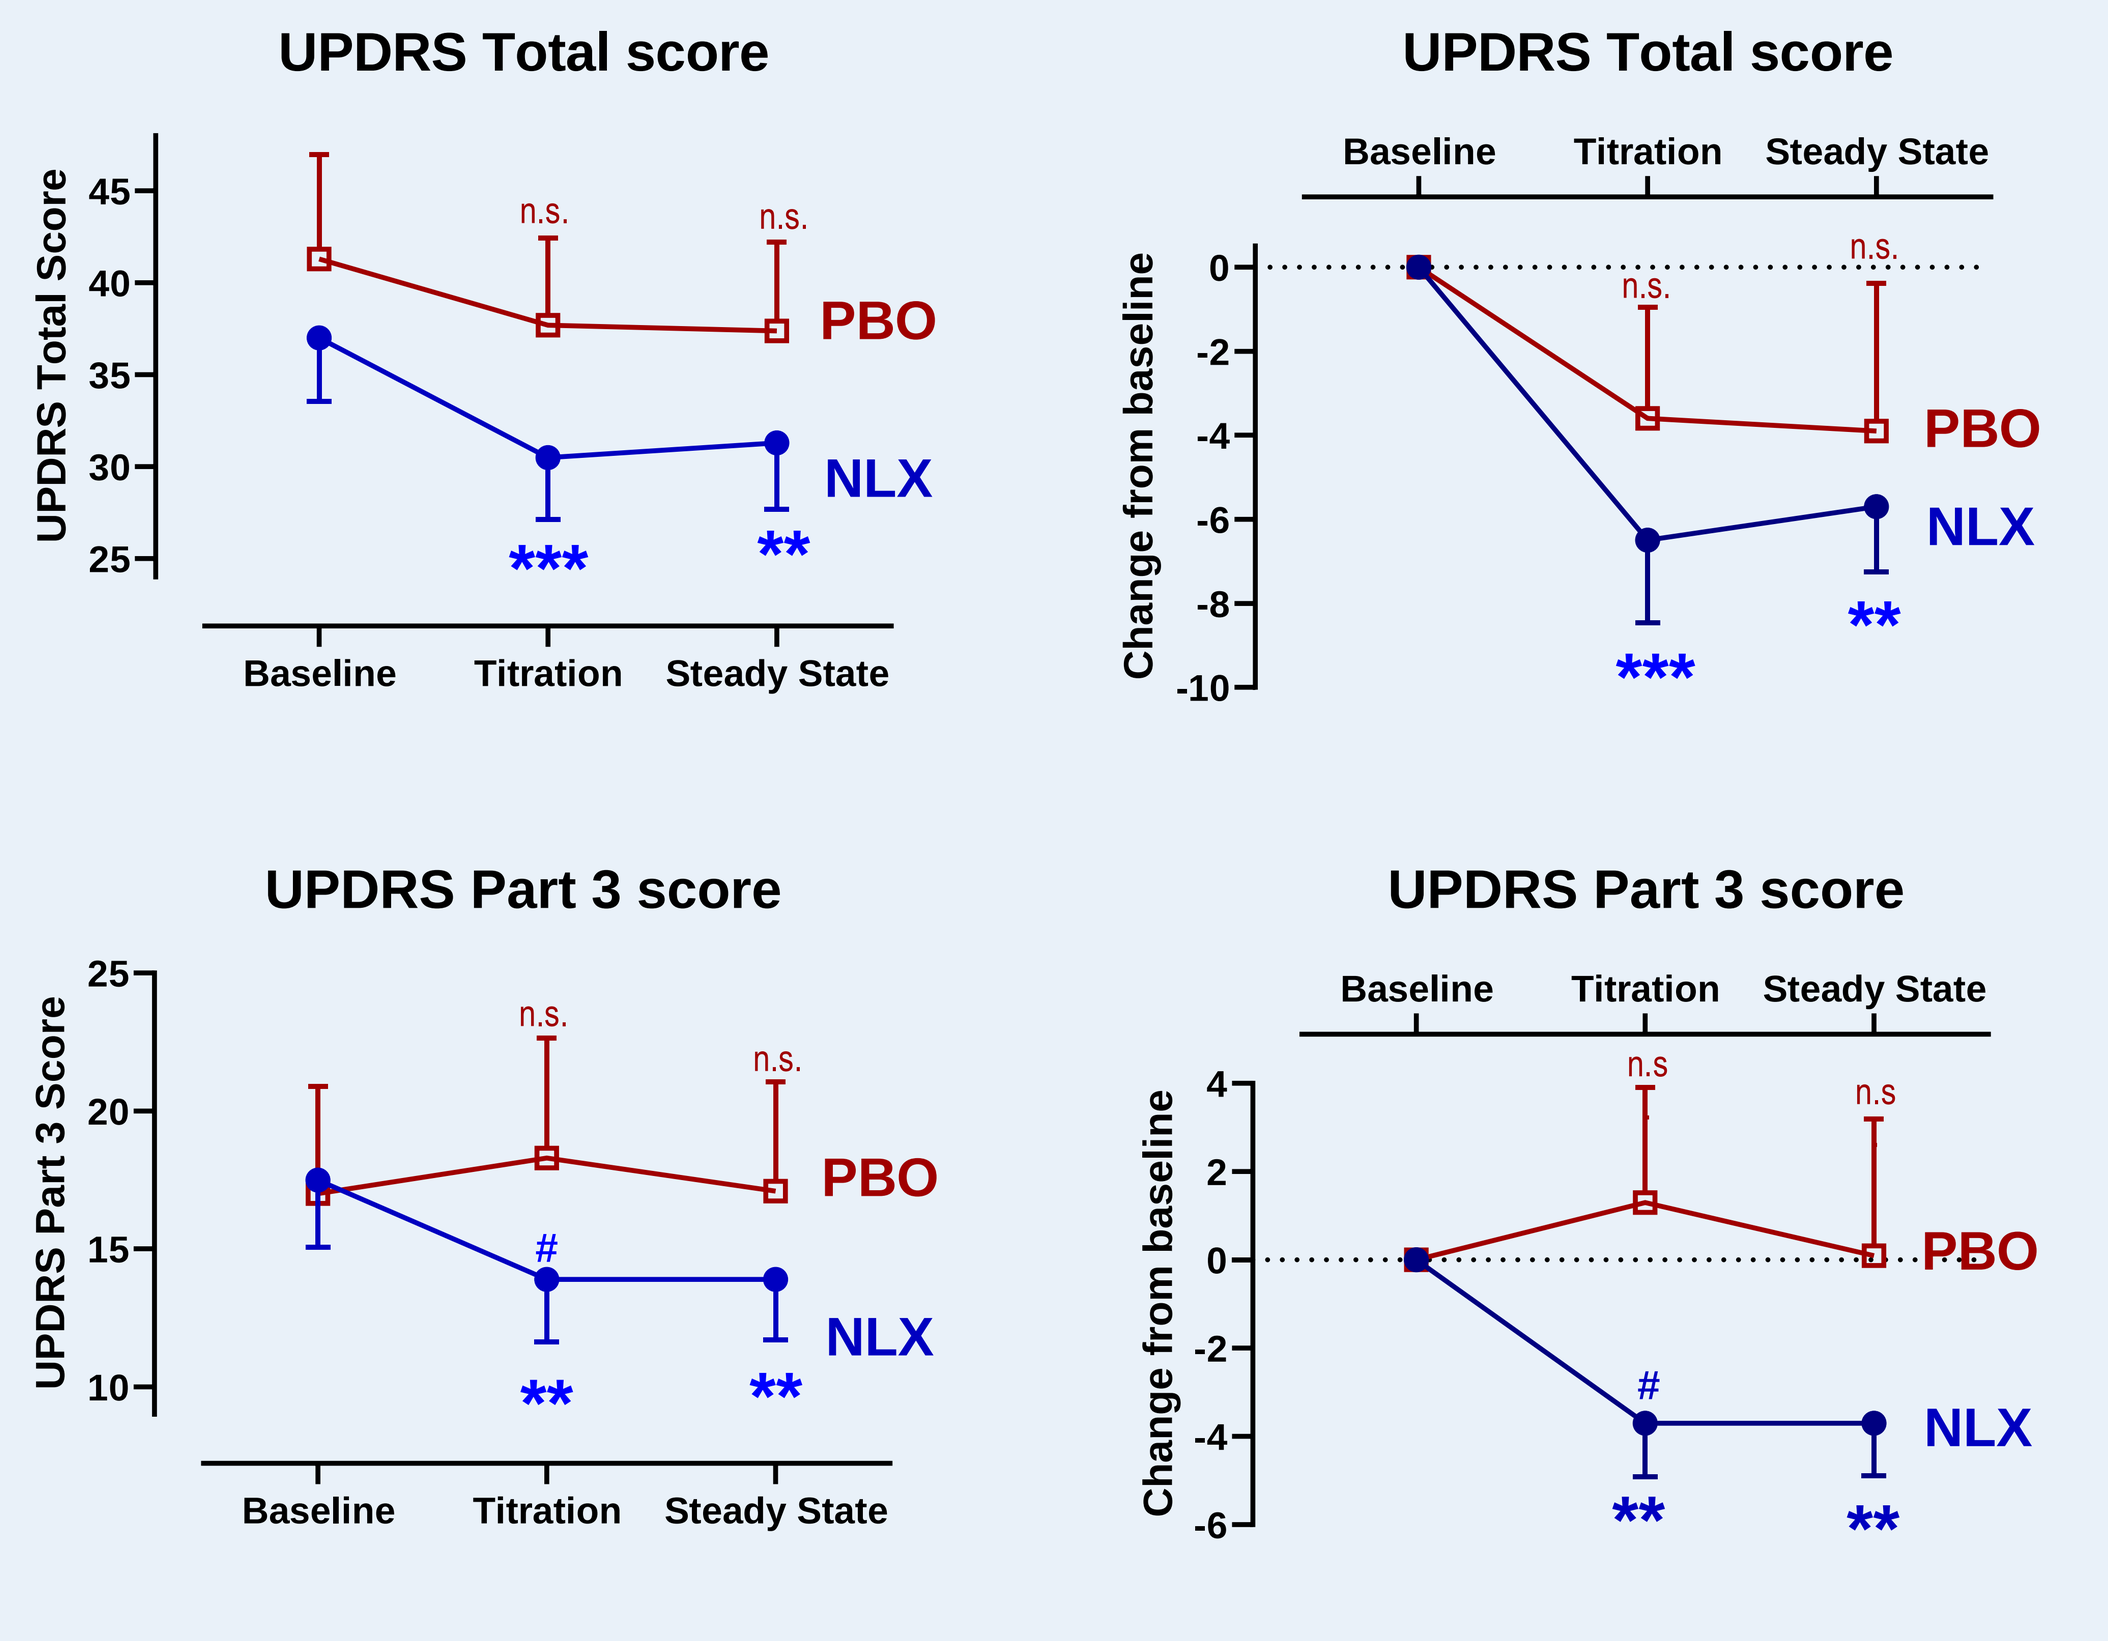
UPDRS Total score
UPDRS Total Score
PBO
NLX
Baseline Titration Steady State
UPDRS Total score
Change from baseline
UPDRS Part 3 score
UPDRS Part 3 Score
UPDRS Part 3 score

At day 28 (end of up-titration), in the NLX-112 group, UPDRS total score (parts 1-4) decreased by -6.5 points ($p=0.0005$) and part 3 score by -3.7 points ($p=0.0072$) compared to baseline; placebo (PBO) group changes were n.s. (-3.6 and +1.3, respectively).

At day 42 (end of steady state period), UPDRS scores were also reduced in the NLX-112 group: total score decreased by 5.7 points ($p=0.0021$) and part 3 score by 3.7 ($p=0.0072$) compared to baseline; PBO group changes were n.s. (-3.9 and +0.1, respectively).

****** p <0.01 vs baseline; ******* p< 0.001 vs baseline; # p<0.05 vs PBO; n.s. not significant

4) NLX-112 improved CGI-C score

Proportions of subjects showing changes in CGI-C score at the end of steady state

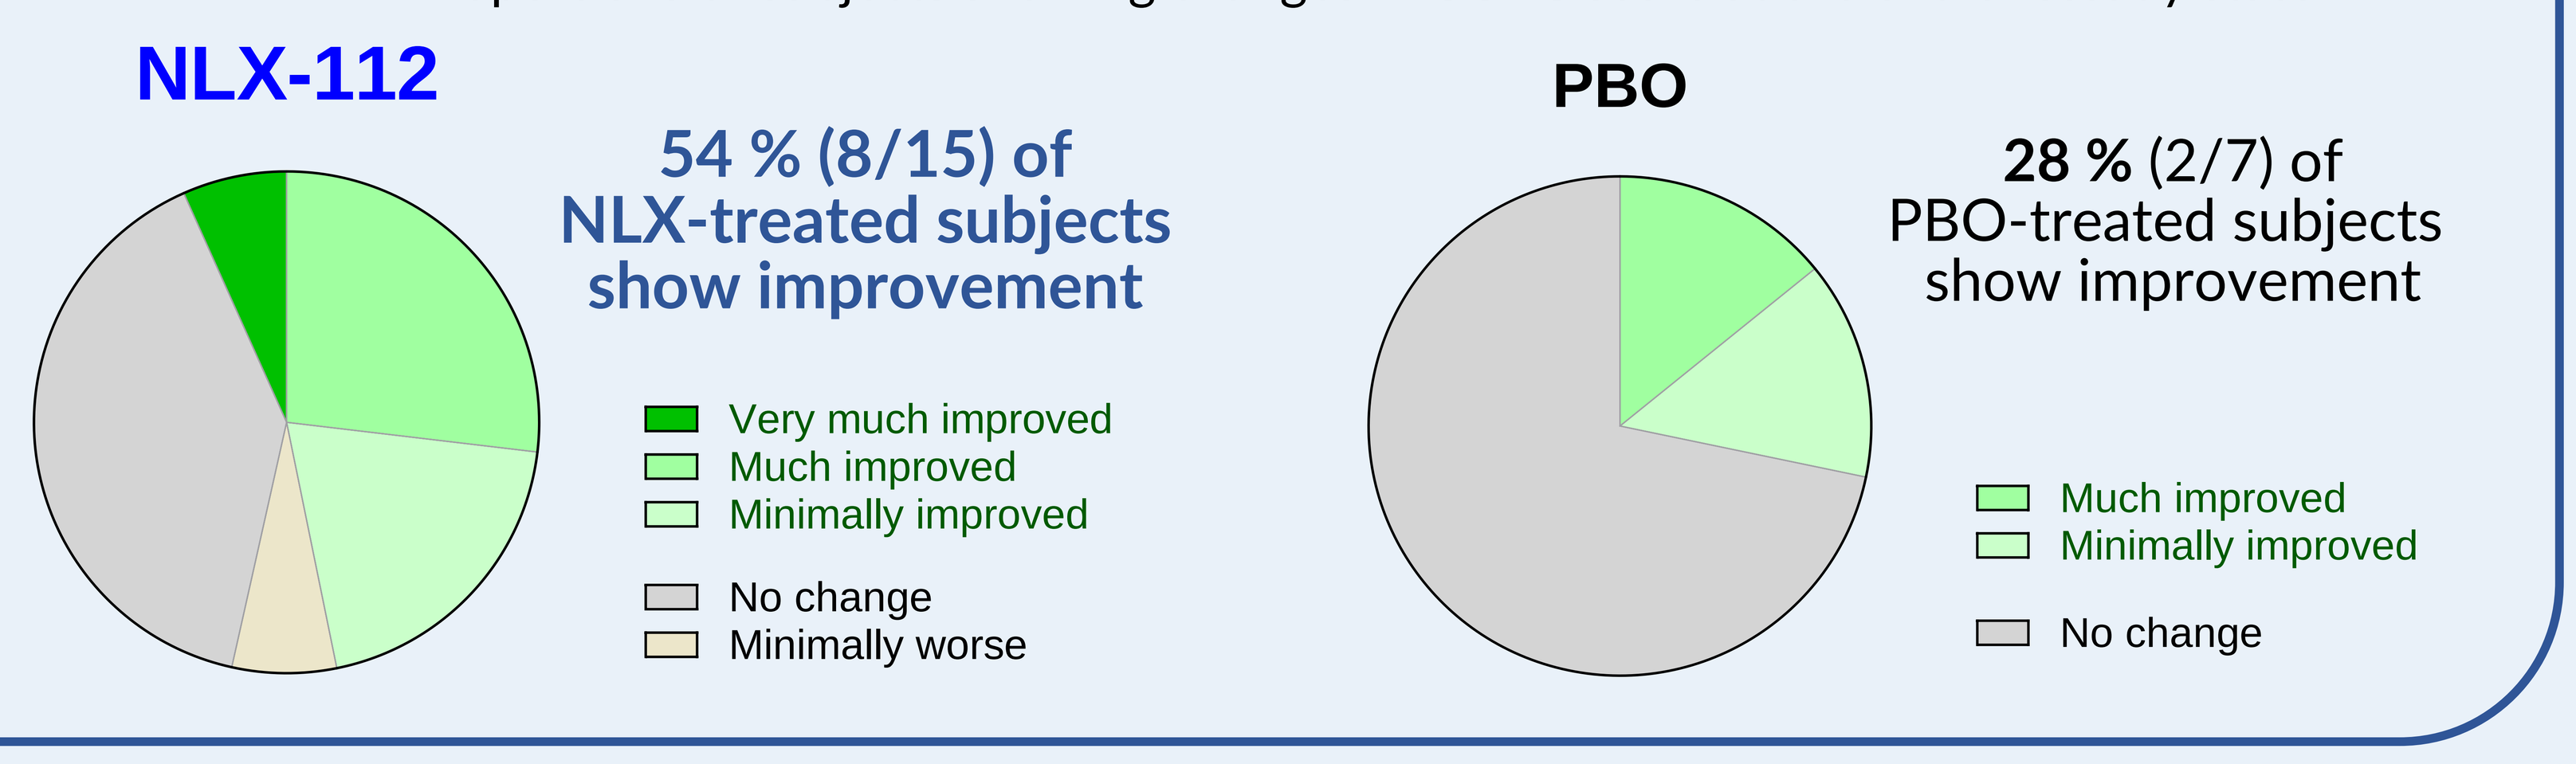
NLX-112
54 % (8/15) of NLX-treated subjects show improvement
Very much improved
Much improved
Minimally improved
No change
Minimally worse
PBO
28 % (2/7) of PBO-treated subjects show improvement
Much improved
Minimally improved
No change

Summary and conclusions

1) In this population of PwP exhibiting moderate to severe LID, NLX-112 was safe and well tolerated in most participants.
2) NLX-112 significantly reduced LID (as measured by UDysRS) in a treatment-duration dependent manner.
3) Unlike previous observations with poorly-selective serotonin 5-HT$_{1A}$ agonists, the anti-LID activity of NLX-112 did not interfere with the antiparkinsonian activity of levodopa. Instead, NLX-112 elicited a further reduction of parkinsonism (as measured by UPDRS).
4) More PwP on NLX-112 than on placebo showed improvement in CGI-C.

➔ **This is the first example of a drug that targets the 5-HT system and shows <u>both anti-LID and anti-parkinsonian activity</u>.**

➔ **NLX-112 could constitute a first-in-class dual-acting treatment for movement symptoms in PwP.**

References

1. Newman-Tancredi, A., et al., J Pharm Pharmacol, 2017. 69(9): p. 1178-1190.
2. Iderberg, H., et al., Exp Neurol, 2015. 271: p. 335-350.
3. McCreary A., et al., Neuropharmacology, 2016.;105: p .651-660.
4. Fisher, R., et al., Neuropharmacology, 2020. 167: p. 107997.
5. Depoortere, R., et al., Parkinsonism Relat Disord, 2020. 78: p. 151-157.

Movement Disorders Society congress, August 27-31, 2023, Copenhagen, Denmark. Abstract #1435